Abstract number: 4CPS-100, ATC code: J06 - Immune sera and immunoglobulins

QUALITY OF INTRATECT: IN VITRO EVALUATION OF BIOLOGICAL ACTIVITIES

C. SCHMIDT, K. WINTERLING, C.C. HEINZ, M. KÖNIG, V. BRAUN,
S. KISTNER, M. GERMER, BIOTEST AG, PRECLINICAL RESEARCH

From Nature for Life

Background

Human normal immunoglobulin preparations for intravenous application (IVIG) such as Biotest's Intratect were initially developed as replacement therapy for primary and secondary immuno-deficiencies (PID, SID). Over the time the clinical use has broadened and includes the treatment of patients with autoimmune diseases. This hyperinflammatory conditions are typically treated at higher doses (Wahn, 2016). Recently, Chronic Inflammatory Demyelinating Polyradi-culo¬neuropathy (CIDP) and multifocal motor neuropasthy (MMN) have become formally approved indications.

Methods

Material

Consecutive recent batches of the Intratect product family (Intratect, Zutectra, Fovepta, Hepatect CP, Varitect CP, Cytotect CP Biotest) were analysed for safety relevant parameters. Functional immunological activities were analysed in three batches of Intratect and two other IVIG brands (Privigen, Kiovig).

Safety relevant tests

Since thrombogenicity is a general risk identified for IVIGs, analyses of Intratect with globally-used coagulation tests, such as the thrombin generation assay, were performed. Additionally, specific tests for the detection of potential impurities (e.g. prekallikrein activator [PKA]) were employed to assess the thrombogenic potential.

The tests for anti-A and anti-B hemagglutinins complies with the European Pharmacopoia (2.6.20).

Anticomplementary activity was determined by a complement consumption method recommended by the European Pharmacopoeia.

Immunological assays

EBV-specific IgG antibody levels were determined using VCA IgG specific EIA (r-biopharm) according to the manufacture's recommendation. IgG values were expressed as arbitrary units (U/ml). All other antigen specific IgG antibodies were determined using commercial diagnostic assays.

Discussion and Conclusions

- Intratect was found to be free of procoagulant and other impurities.
- The content of blood group antibodies, which are associated with the risk of hemolysis, can be controlled by the manufacturing process. In Intratect, these antibodies are consistently tightly controlled and their content is far below the isoagglutinin titer threshold.
- Pathogen antigen recognition is a prerequisite for the anti-infective activity of immunoglobulins. Intratect was found to contain antibody titers against relevant viruses and bacteria.
- Quality characteristics of IVIG preparations differ from brand to brand but are typically consistent from batch to batch for a single brand.
- Multiple factors contribute to the quality of the IVIG preparations. Important quality attributes are associated with safety (Bellac et al. 2015, Etscheid et al. 2012), and adequate antimicrobial activity. Different manufacturing processes determine differences in quality, safety and efficacy of IVIG brands.

References

- Bellac CL et al., The role of isoagglutinins in intravenous immunoglobulin-related hemolysis.Transfusion. 2015;55 Suppl 2:S13-22
- Etscheid M et al., Identification of kallikrein and FXIa as impurities in therapeutic immunoglobulins: implications for the safety and control of intravenous blood products. Vox Sang. 2012 Jan;102(1):40-6
- Wahn V, From immune substitution to immunomodulation, Semin Hematol. 2016;53 Suppl 1:S7-9

Carolin Schmidt and Karina Winterling contributed equally to this work.

Purpose

Due to this change in use which involves increased doses, and in view of adverse events observed with other IVIG brands, we have reevaluated quality parameters relevant for product safety and efficacy.

Results

Commercial IVIG preparations contain controlled amounts of antibodies against the blood group antigens A and B. The hemagglutination assay according to the European Pharmacopoia demonstrated consistent low titers in Intratect.

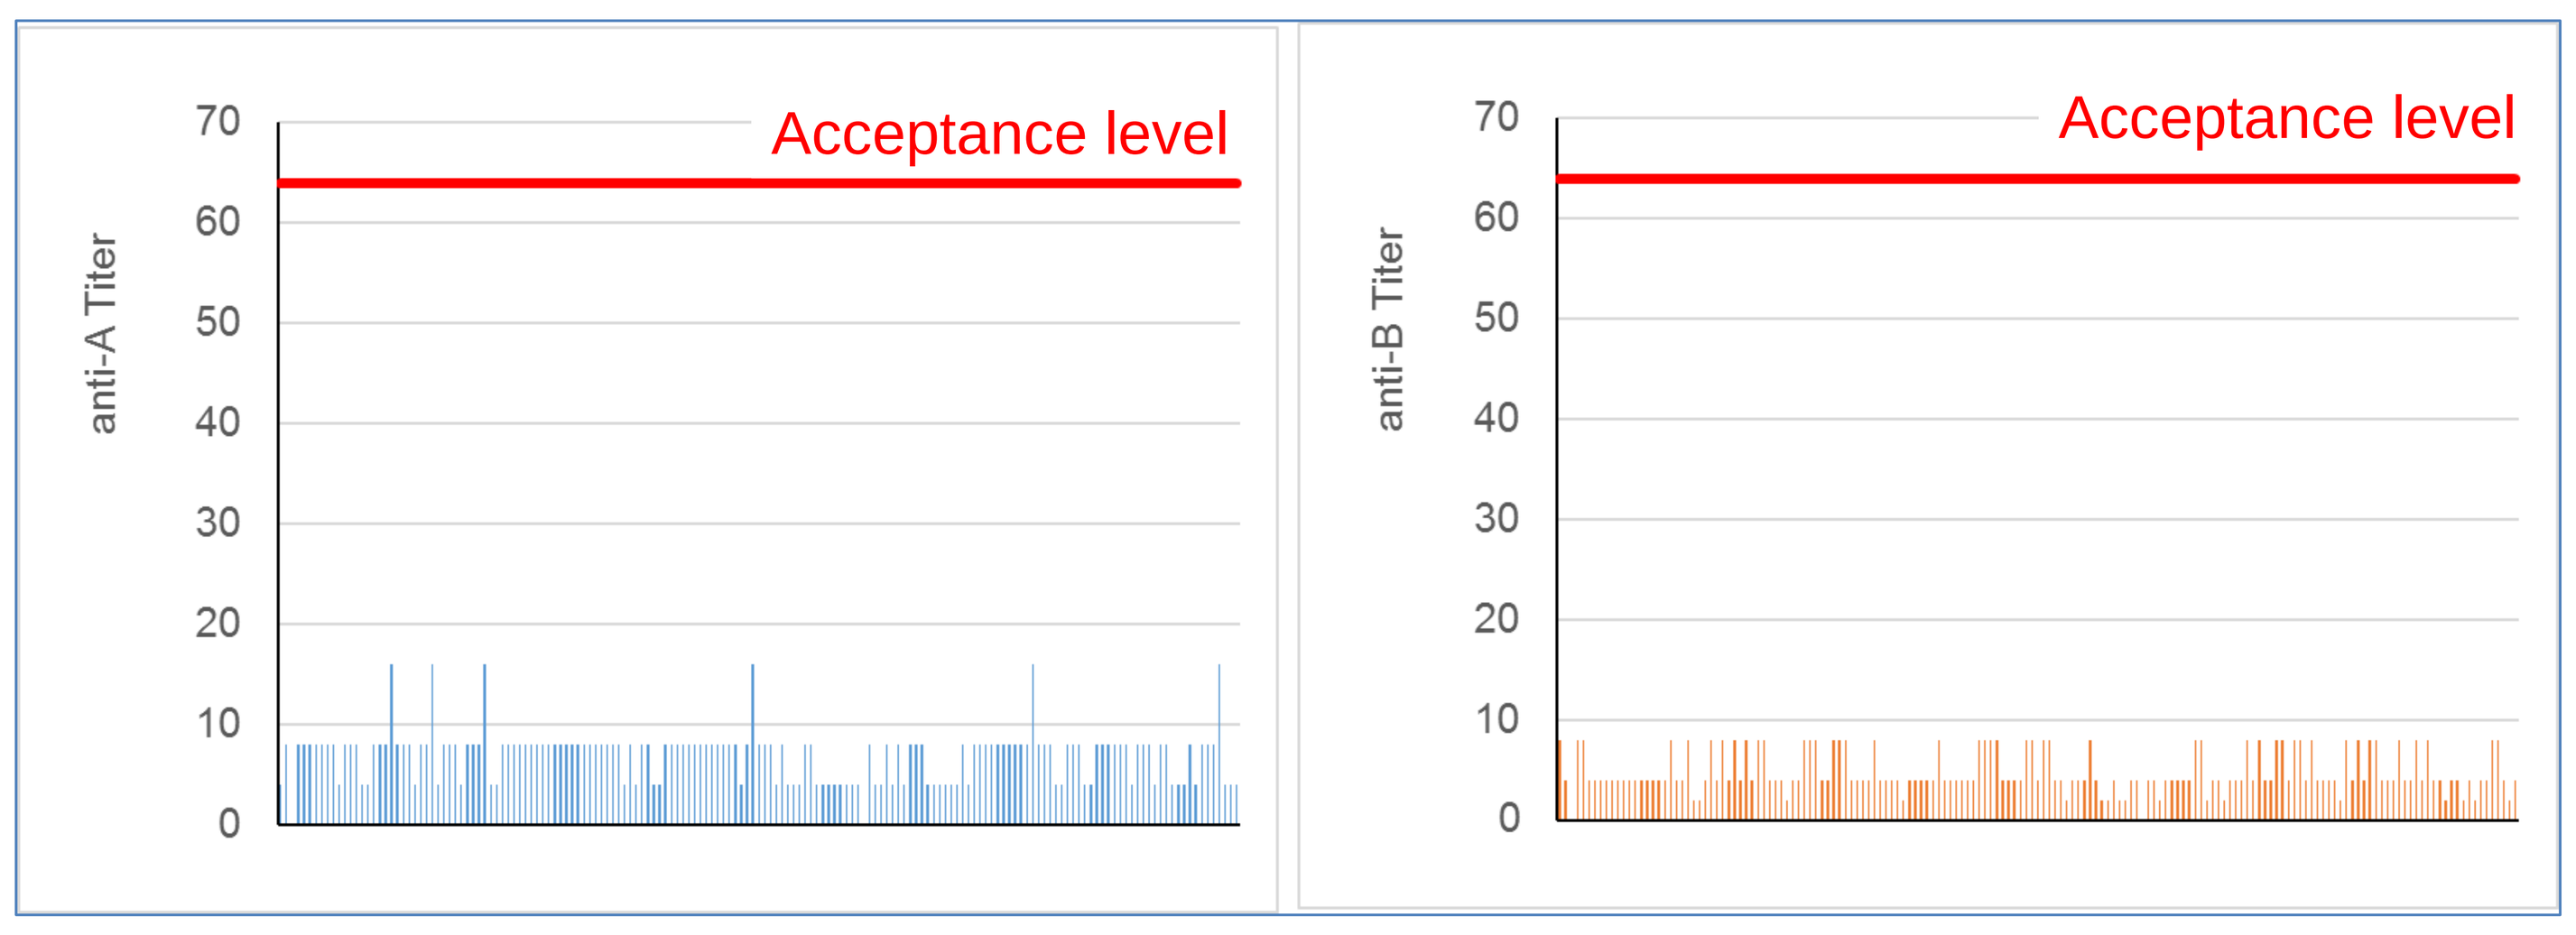
anti-A Titer
Acceptance level
anti-B Titer
Acceptance level
0
10
20
30
40
50
60
70

High anticomplementary activity (ACA) may be correlated with a high content of large size polymers in IVIG and may lead to adverse reactions. It should be lower than 1.0 CH50/mg protein. In Intratect, the variability of ACA is low and in a narrow range.

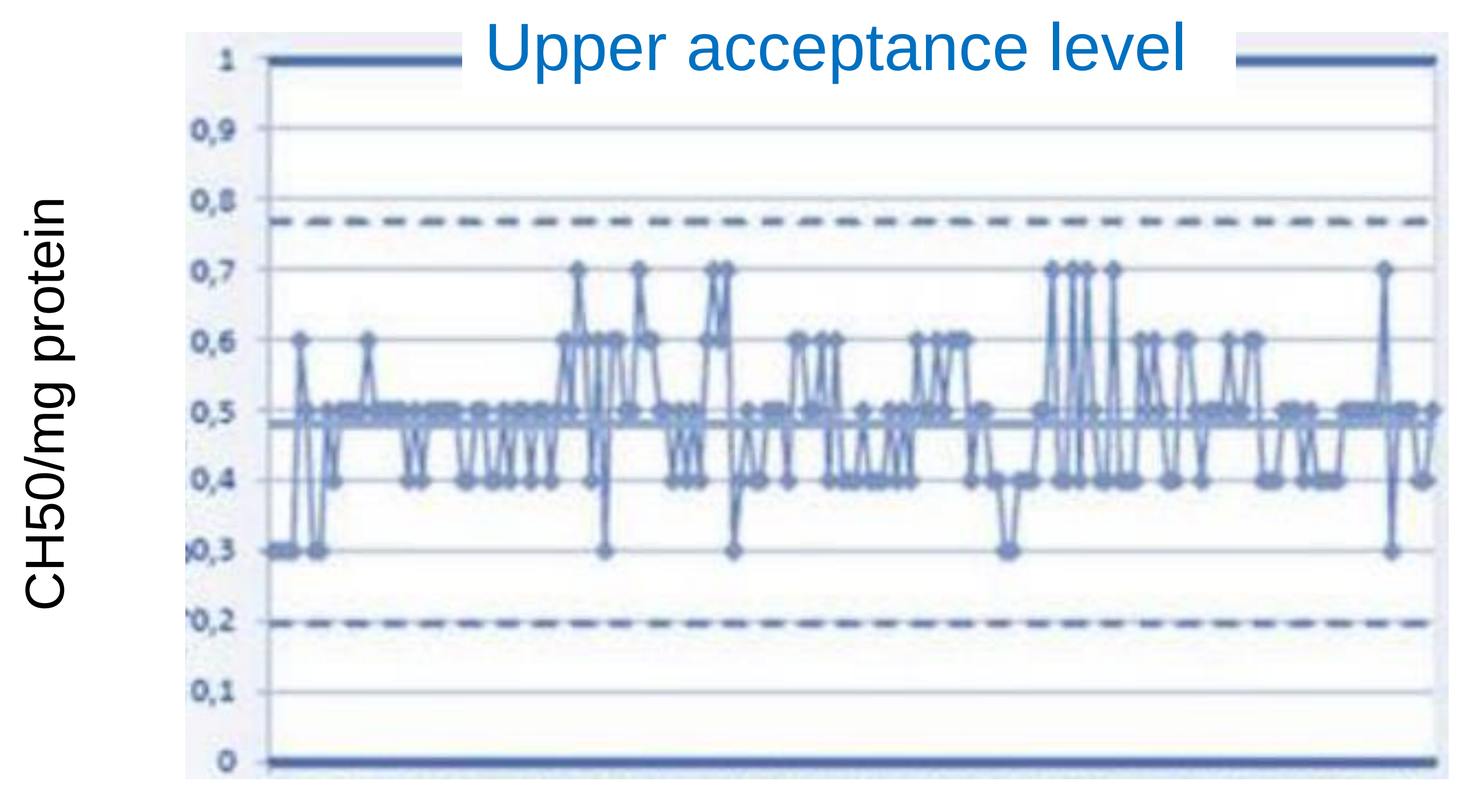
CH50/mg protein
Upper acceptance level
0
0,1
0,2
0,3
0,4
0,5
0,6
0,7
0,8
0,9
1

As a general test for thrombogenic impurities a thrombin generation assay was performed. All batches contained undetectable levels of procoagulant activity.

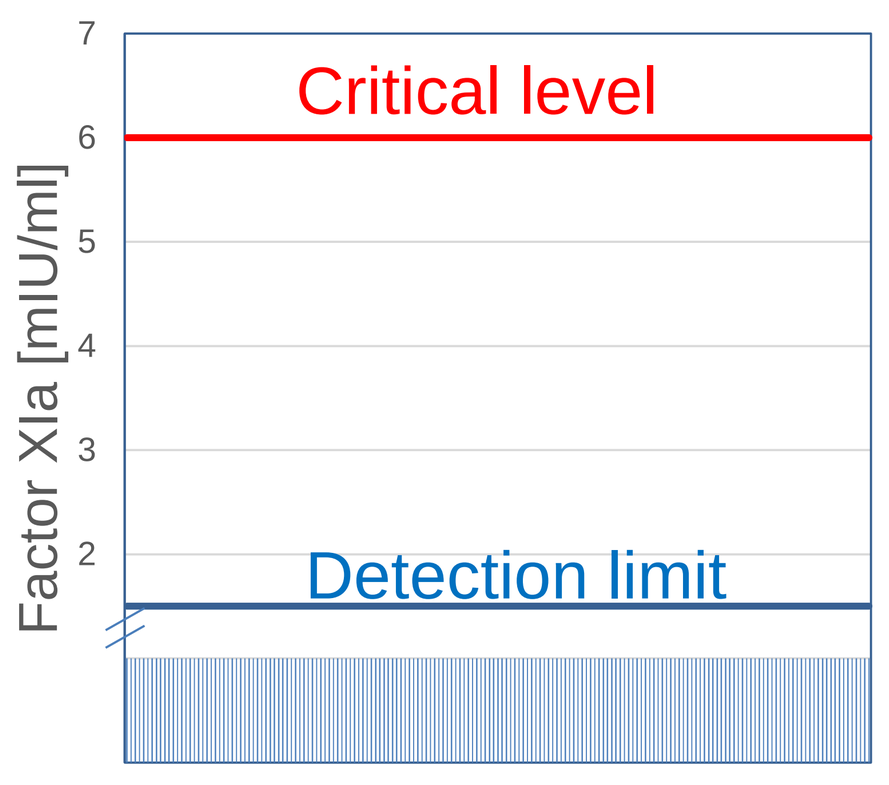
Factor XIa [mIU/ml]
Critical level
Detection limit
2
3
4
5
6
7

The content of antibodies to Toxoplasma gondii and hepatitis E virus was highest in Intratect. Some differences can be attributed to the predominant plasma source (e.g. West Nile virus and Zika virus antibodies in Intratect [predominantly EU plasma] vs. comparator products [higher content of US plasma]). The content of antibodies to a number of common pathogens was comparable in all tested IVIG products.

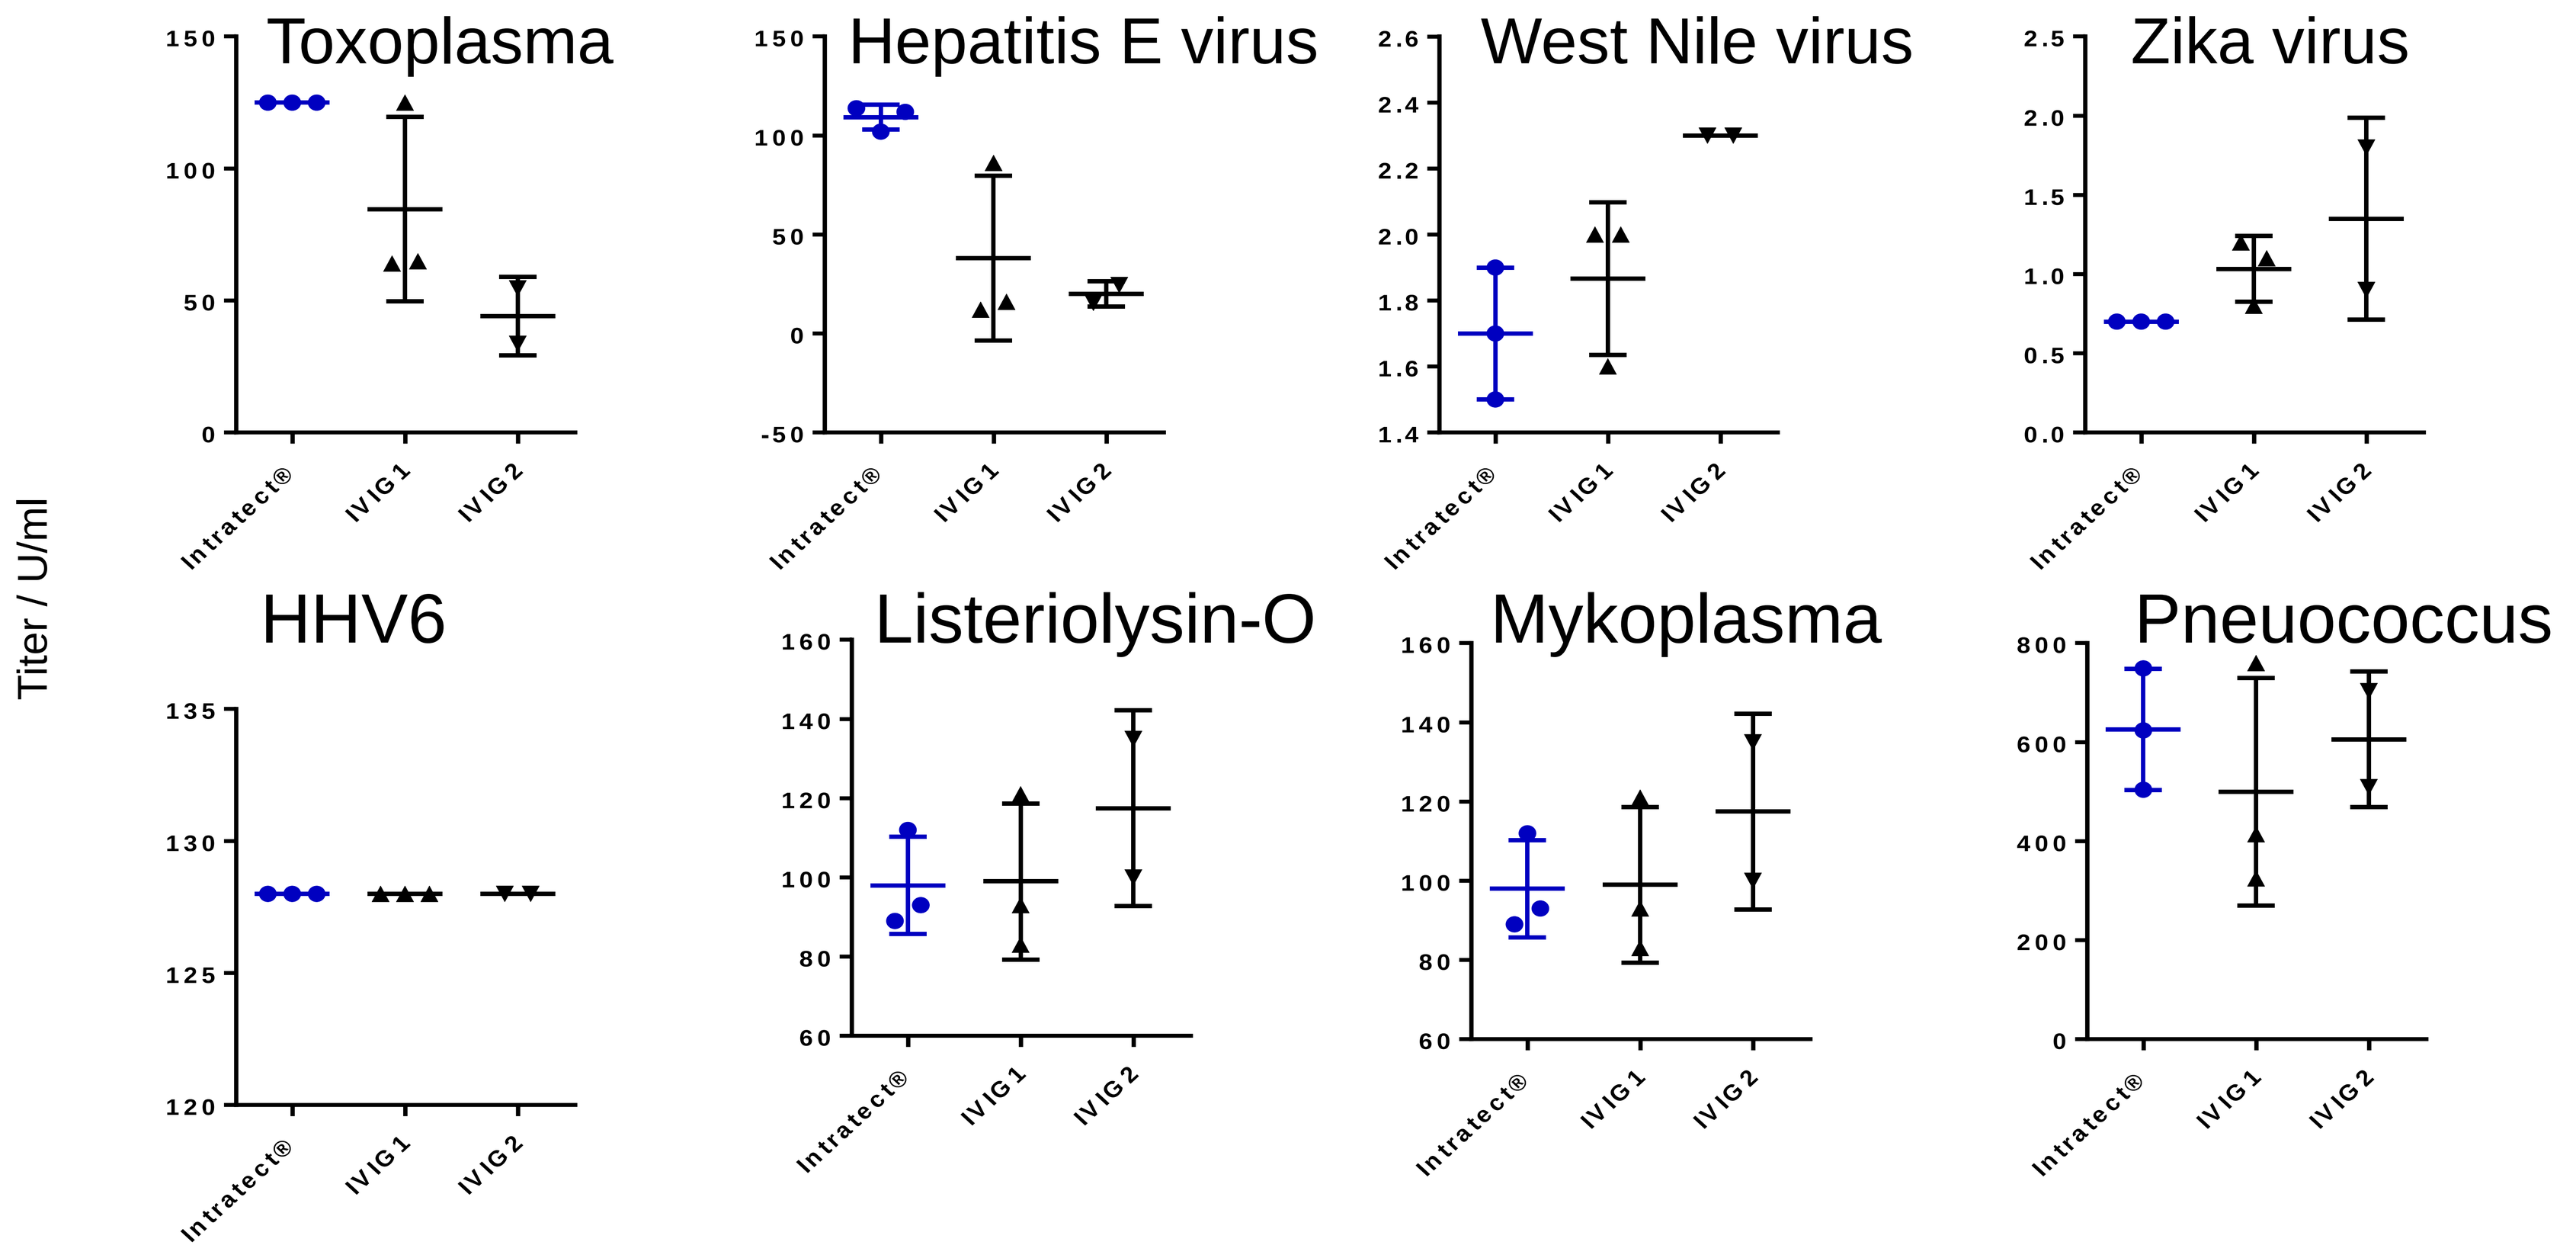
Titer / U/ml
Toxoplasma
Hepatitis E virus
West Nile virus
Zika virus
HHV6
Listeriolysin-O
Mykoplasma
Pneuococcus
Intratect®
IVIG1
IVIG2

Biologically relevant antibodies were consistently present in IVIG preparations. The products differ in the content of some specific antibodies depending on the source material and the production process.

Acknowledgement

The authors thank Prof. Gisela Enders & Kollegen MVZ GbR, Stuttgart, Germany for expert practical assay performance and PD Dr. med. Martin Enders and Dr. Elena Ladwig for excellent organisation of the work and fruitful discussions.

http://www.eahp.eu/24-4CPS-100