KNOWLEDGE, ATTITUDES AND SELF-CARE ACTIVTIES AMONG PATIENTS WITH TYPE II DIABETES

N. Elhatab. 1, J. Silcock. 1, A. Graham.1
1 University of Bradford, Faculty of Life science, School of Pharmacy, Bradford, United Kingdom.

UNIVERSITY of BRADFORD
Faculty of Life Sciences

GM009

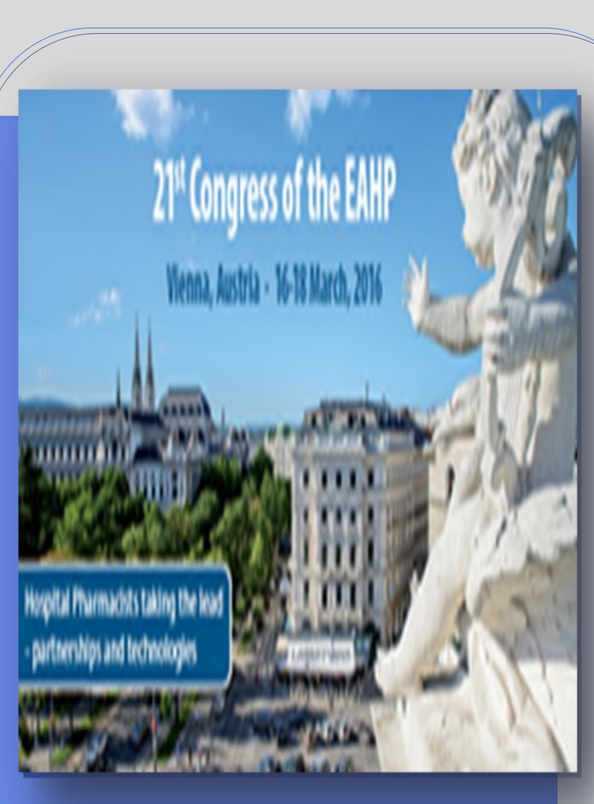
21st Congress of the EAHP
Vienna, Austria - 16-18 March, 2016

Objectives

An association between improved glycaemic control and a sustained decrease in the rate of diabetic complications has been shown in randomized clinical trials (1). This survey aimed to evaluate knowledge, attitudes and self-care activities among patients with type II diabetes in Tripoli, Libya. This was part of baseline assessment in a randomised controlled trial of enhanced pharmaceutical care

Materials and Methods

40 community pharmacies were randomly assigned to be control (18) or intervention (22) premises. Then each pharmacy recruited 4 or 5 type II diabetic patients. Overall, 225 patients were recruited and assigned to receive usual pharmacist care (n=100) or a predefined pharmacist intervention (n=125). Each patient completed a questionnaire in which demographic information, disease history, medication history, diabetes knowledge (2) and self-care activity levels were collected (3). At inclusion patients measured their fasting plasma glucose (FPG) on three consecutive days. The data was analysed by IBM SPSS Statistics 22.

Results

Patients were randomised to control (100, 58% female) and intervention (125, 55% female) groups. There were no baseline differences in mean (± sd) age (56±13 vs. 55±12.3) or FPG (176.88±41.30mg/dl vs 177.93±39.69mg/dl). The baseline FPG is much higher than the normal range of 70-100 mg/dl, which indicates capacity to benefit from better care. Knowledge scores were also similar in the control (6/15±2.5) and intervention groups (6.6/15 ±2.1). There was a positive correlation between (FPG) and the frequency of high fat food (red meat and full fat dairy products) consumption in the previous 7 days (p=0.04). The belief that type II diabetes is a serious problem was positively correlated with higher (FPG) levels (p=0.02) in the intervention group. High levels of (FPG) were positively correlated with the poor diabetes knowledge test scores in the control group (p=0.01).

Table 1: Demographic characteristics

Parameter	Control group (n=100)	Intervention group (n=125)
Male	42% (42)	45% (56)
Female	58%(58)	55%(69)
Age (years)	56±13 (35-80)	55.22±12 (37-85)
Duration of having diabetes	7.7±6.1 (1-21)	7.52±4.88 (1-25)
Education		
No education	6% (6)	6% (8)
Primary school	41% (41)	40% (50)
Secondary school	33% (33)	32% (40)
High education (i.e. BSc, MSc)	20% (20)	22% (27)
Smokers	12% (12)	18% (23)
People manage their blood glucose control with:		
One tablet	46% (46)	45% (55)
Two tablets	38% (38)	36% (45)
Three tablets	9% (9)	11% (13)
On diet	4% (4)	3% (4)
Insulin users	3% (3)	5% (6)
FPG (mg/dl)	176.88 ± 41.30 (107.5-372.9)	177.93±39.69 (107.5-372.9)

Table 2: Diabetes Knowledge Test

Questions about:	Control Correct answers	Intervention Correct answers
Diabetes definition	4% (4)	5% (6)
Infection management	11% (11)	22 % (27)
Reasons of doing exercise	16% (16)	20% (25)
Understanding of neuropathy complication	17% (17)	21% (26)
Reasons of test Blood Glucose	24% (24)	27% (34)
Understanding diabetes complication	31% (31)	36% (45)
Understanding eye check-ups	33% (33)	34% (43)
Understanding of diet plan	46% (46)	48% (60)
Recording BG measurements	47% (47)	63% (79)
Medication checks	50% (50)	52% (65)
Normal range of BG postprandial	51% (51)	52% (65)
Frequency of exercise	60% (60)	61% (76)
Hypoglycaemic management	65% (65)	66% (83)
Normal range of BG pre-prandial	73% (73)	74% (93)
Practical Method of test BG	88% (88)	87% (109)

Table 3: Diabetes Self care Activities

Frequency of activities during last seven days measured by scale (0-7 days)	Average scale (control)	Average Scale (Intervention)
Specific exercise session (such as swimming, walking, biking) other than what you do around the house or as part of you work	1.8±2.1	2.2±2.2
Eating plan?	2.1±1.7	2.6±2.1
Exercise at least 30 minutes of physical activity? activity, including walking	2.4±2.2	2.6±2.0
Space carbohydrates evenly through the day	2.7±2.0	2.5±1.9
Healthy eating plan	2.7±2.1	2.5±2.1
Eat high fat foods such as red meat or full fat dairy products	2.5±1.8	2.4±1.8
Test your blood sugar the number of times recommended by your health care provider	2.5±1.9	2.5±1.9
Test your blood sugar?	3.0±2.0	2.8±1.9
Check your feet?	3.1±2.3	3.2±2.3
Inspect the inside of your shoes?	3.3±2.3	3.4±2.3
Eat five or more servings of fruits and vegetables	3.4±2.1	3.5±1.9

Discussion

Our data shows that the poor glycaemic control among participants may be associated with lower levels of diabetes knowledge and poor self-care behaviour. Since patients believe that type II diabetes is serious disease, they understand the risks but have difficulty taking action to control their blood glucose and avoid diabetes complications. This preliminary finding is consistent with previous studies (4)

Conclusion

This baseline data shows poor diabetes knowledge and high levels of FPG in both control and intervention groups. Glycaemic control might be improved if enhanced pharmaceutical care can change patients' attitudes towards diabetes or eating habits, and enhance self-efficacy.

References

1. Mehuys.L, Bortle.V, DeBolle.L, Tongelen,V, Annemans.L, Remon.P, Giris.M. . Effectivnes of a community pharm,aciust intervemntion in diabetes care: a randomised controlled trial.. Journal oif Cxlinical Pharmcy and Therapeutics 2011; 36(): .
2. Egenmann,C, Skinner.T, Colagiuri.R. Development and validation of a diabetes knowledge questionnaires. . Practical diabetes international 2011; 28(4): .
3. Toobert DJ, Hampson SE, Glasgow RE . The summary of diabetes self-care activities measure: results from 7 studies and a revised scale. . Practical diabetes international 2000; 23(): 943–950.
4. Daly,T.M, Hartz, A.J, Xu, Y, Levy, B.T, James, P.A, Merchant, M.L, Garrett, R.E . An Assessment of Attitudes, Behaviours, and Outcomes of Patients with Type 2 Diabetes. Journal of American Board Family Medicine 2009; 22(3): .

Acknowledgement

I would like to express my sincere gratitude to my supervisors Dr Jon Silcock & Dr Anne Graham for the continuous support of my PhD study and my husband and my family.