COMPARISON OF ROBOTIC AND MANUAL RECONSTITUTION: STABILITY ASSESSMENT OF PROTEIN DRUGS IN HOSPITAL COMPOUNDING

I. CEDERWALL[A], C. SABATÉ MARTÍNEZ[B] U. ELOFSSON[A], A. MILLQVIST FUREBY[A,E], R. BAGHDARSARIAN NAMAGERDI[C], B. FURTMANN[D], C. SCHOENAU[D], M. WAHLGREN[E] AND M. PAULSSON[B]*

[A]RISE Research Institute of Sweden, Stockholm, Sweden, [B]Department of Women's and Children's Health, Uppsala University, Akademiska sjukhuset, SE-751 85 Uppsala, Sweden, [C]Hospital Pharmacy Compouding unit, Uppsala, Sweden, [D]Sanofi-Aventis Deutschland GmbH, Biologics Drug Product Development & Manufacturing, Industriepark Hoechst, K703. Brüningstr. 50, 65926 Frankfurt am Main, Germany, [E]Department of Food Technology, Lund University, P.O. Box 124, 22100 Lund, Sweden . *mattias.paulsson@uu.se

3PC-020

Background and Importance

- Protein drugs are widely used in hospitals for treating cancer and immune diseases
- Manual reconstitution introduces risks of human error, variability, and handling stress on proteins[1]
- Silicone oil can be introduced from syringes or closed system transfer device

Automated reconstitution offers improved safety, and reduced workload. Studies suggest that robotic compounding provides comparable/improved quality compared to manual preparation[2]

Why is this important for hospital pharmacists?
✓ Reduces **risk of protein degradation**
✓ Supports **better patient safety**

Aim and Objectives

Aim: To evaluate automated vs. manual reconstitution of protein drugs and assess their impact on drug stability
Objectives: Compare **five reconstitution methods** (manual vs. four robotic programs)

Materials and Methods

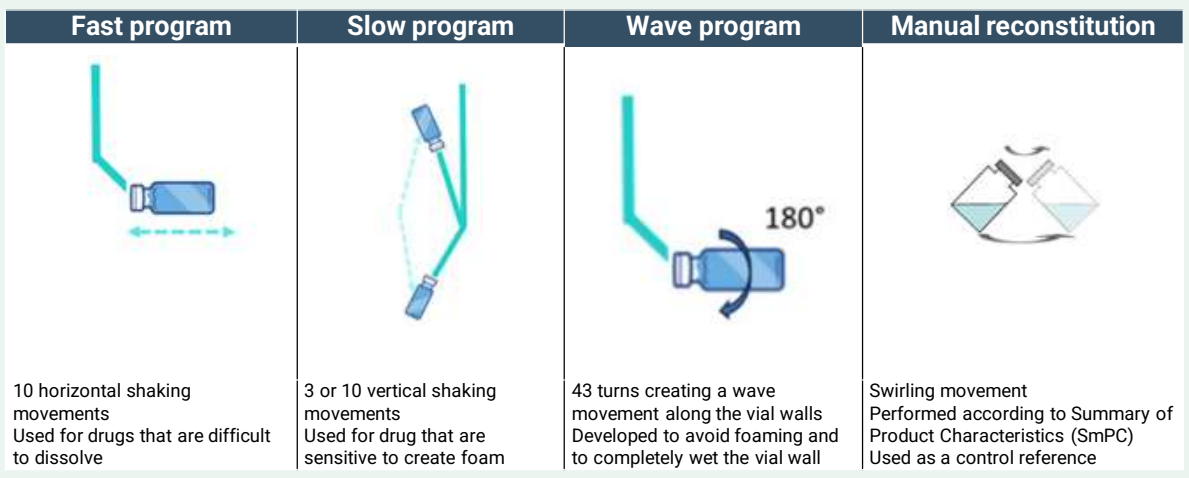
Fast program
10 horizontal shaking movements
Used for drugs that are difficult to dissolve
Slow program
3 or 10 vertical shaking movements
Used for drug that are sensitive to create foam
Wave program
180°
43 turns creating a wave movement along the vial walls
Developed to avoid foaming and to completely wet the vial wall
Manual reconstitution
Swirling movement
Performed according to Summary of Product Characteristics (SmPC)
Used as a control reference

Protein drugs tested (supplied by EFPIA partners):
- **A** (Nanobody, liquid)
- **B** (Bispecific antibody, lyophilized)
- **C** (Nanobody, lyophilized)
- **D** (Bispecific antibody, lyophilized)

Reconstitution methods compared:
✓ **Manual reconstitution** (Uppsala, Barcelona)
✓ **Robotic** Grifols Kiro Oncology (Fast, Slow3/10, Wave)

Analyses performed:
-> Visual inspection for foaming & air bubbles
-> Dynamic Light Scattering (DLS) for for nanometer particle distribution
-> Flow Imaging Microscopy for subvisible particles
-> Size-Exclusion Chromatography for aggregate formation and monomer loss

Results

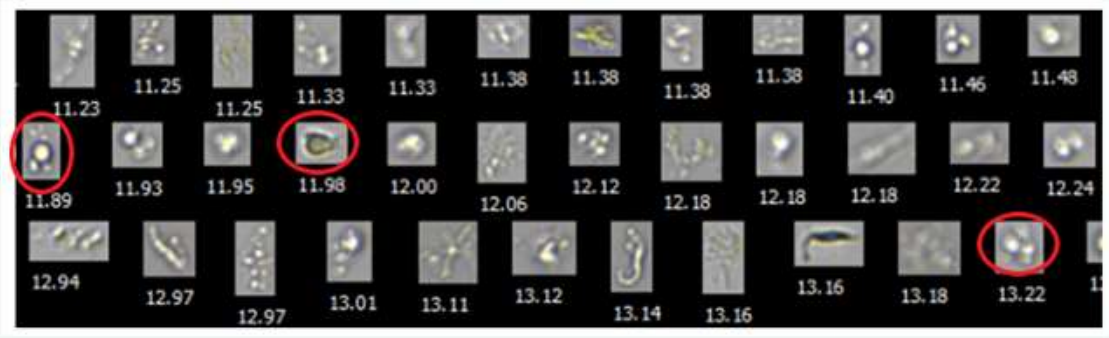
11.23
11.25
11.25
11.33
11.33
11.38
11.38
11.38
11.38
11.40
11.46
11.48
11.89
11.93
11.95
11.98
12.00
12.06
12.12
12.18
12.18
12.18
12.22
12.24
12.94
12.97
12.97
13.01
13.11
13.12
13.14
13.16
13.16
13.18
13.22

Fig. 1 Flowcam particle images of protein D fast program. Protein-SO complexes in red circles. Area based diameter (μm) in white.

Particle Formation:
- No clear advantage of one robotic method over another.
- Protein characteristics (formulation, vial design) mattered more than the reconstitution method

Stability:
- No major impact on protein aggregate formation and monomer loss was seen across different methods
- Wave-based reconstitution showed slightly gentler handling

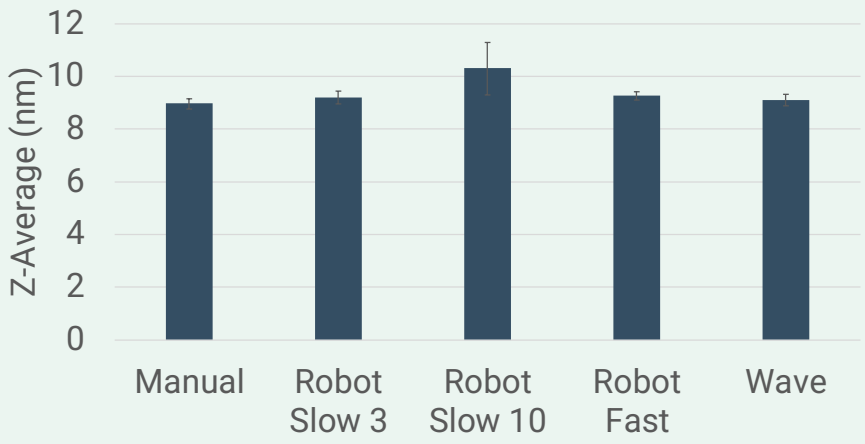
Z-Average (nm)
0
2
4
6
8
10
12
Manual
Robot Slow 3
Robot Slow 10
Robot Fast
Wave

Fig. 2
DLS Z-average of ProteinD reconstituted using different protocols. n=3

For challenging drug formulations, where lyophilizates form a gel-like cake on the vial wall, verifying dissolution without visual inspection is challenging. Fully automated robotic systems may struggle with these formulations

Conclusion and Relevance

✓ Automated reconstitution is a viable alternative to manual preparation in hospital settings
✓ Protein formulation and vial design impact stability more than reconstitution method. The volume of the injection solution relative to the container may cause sloshing, leading to air entrainment and potential hydrodynamic stress on the drug solution previously seen in syringes [3]
✓ Silicone oil can be introduced from siliconized rubber membranes during IV preparation. Keeping vials upright minimizes this risk manually but is not feasible in robotic systems. However, this contribution may be negligible compared to the syringe plunger and CSTD

What does this mean for hospital pharmacists?
◆ Robotics may improve safety and efficiency in aseptic drug preparation
◆ Possibility to choose reconstitution methods based on protein formulation factors
◆ Further optimization and training are required for robotic systems in hospital pharmacies

References and Acknowledgements

1. Cappelletto et al. J Pharm Sci 2024 DOI 10.1016/j.xphs.2024.05.027
2. Geersing et al. 2023 J Pharm Sci DOI 10.1016/j.xphs.2023.10.015
3. Eshraghi et al. Int J Pharm 2022 DOI 10.1016/j.ijpharm.2022.122210
This work has received support from the EU/EFPIA Innovative Medicines Initiative 2 Joint Undertaking (RealHOPE grant n° 101007939). Content of this poster reflects only the author's view and the JU is not responsible for any use that may be made of the information it contains.

RealHOPE
Real World Handling of Protein Drugs – Exploration, Evaluation & Education