Abstract 4CPS-098

ADHERENCE: Observational study to assess guselkumab adherence in clinical practice after 12 months of treatment in patients with moderate-to-severe psoriasis

J. Borrás-Blasco[1], N. Pagès-Puigdemont[2], L. Domínguez Senín[3], M. Gómez Zamora[4], G. Mercadal Orfila[5], J.J. Ortiz de Urbina[6], D. Martínez-López[7], E. Ramírez[8].

[1]H. de Sagunto, Valencia, Spain. [2]H. de la Santa Creu i Sant Pau, Barcelona, Spain. [3]H. Juan Ramón Jiménez, Huelva, Spain. [4]H. Son Espases, Islas Baleares, Spain. [5]H. Mateu Orfila, Islas Baleares, Spain. [6]H. de León, León, Spain. [7]Janssen-Cilag, Madrid, Spain. [8]H. La Princesa, Madrid, Spain

Key message: Our interim analysis suggest **adherent patients** have **lower perception of symptoms** and **less psoriasis signs** compared to non-adherent patients.

Background and Importance

- **Discrepancies** between clinical remission and patients' perceptions of their symptoms **are common in psoriasis**. While clinical scores such as psoriasis area severity index (PASI) provide **objective measures** of disease activity, they may **not fully capture** the **patient experience**.
- **Integrating patient-reported outcomes (PROs)** is essential to complement clinical assessments and monitor **treatment effectiveness**.
- **Adherence** to **biologic therapies** is a key factor influencing long-term outcomes, yet its **relationship** with **PROs remains underexplored** in real-world settings.

Aim and Objectives

- This study primarily aimed to assess 12-month **adherence to guselkumab** in patients **with moderate-to-severe psoriasis** in **routine** clinical practice, in Spain. Secondary objectives included exploring the association **between adherence** and **PROs**.

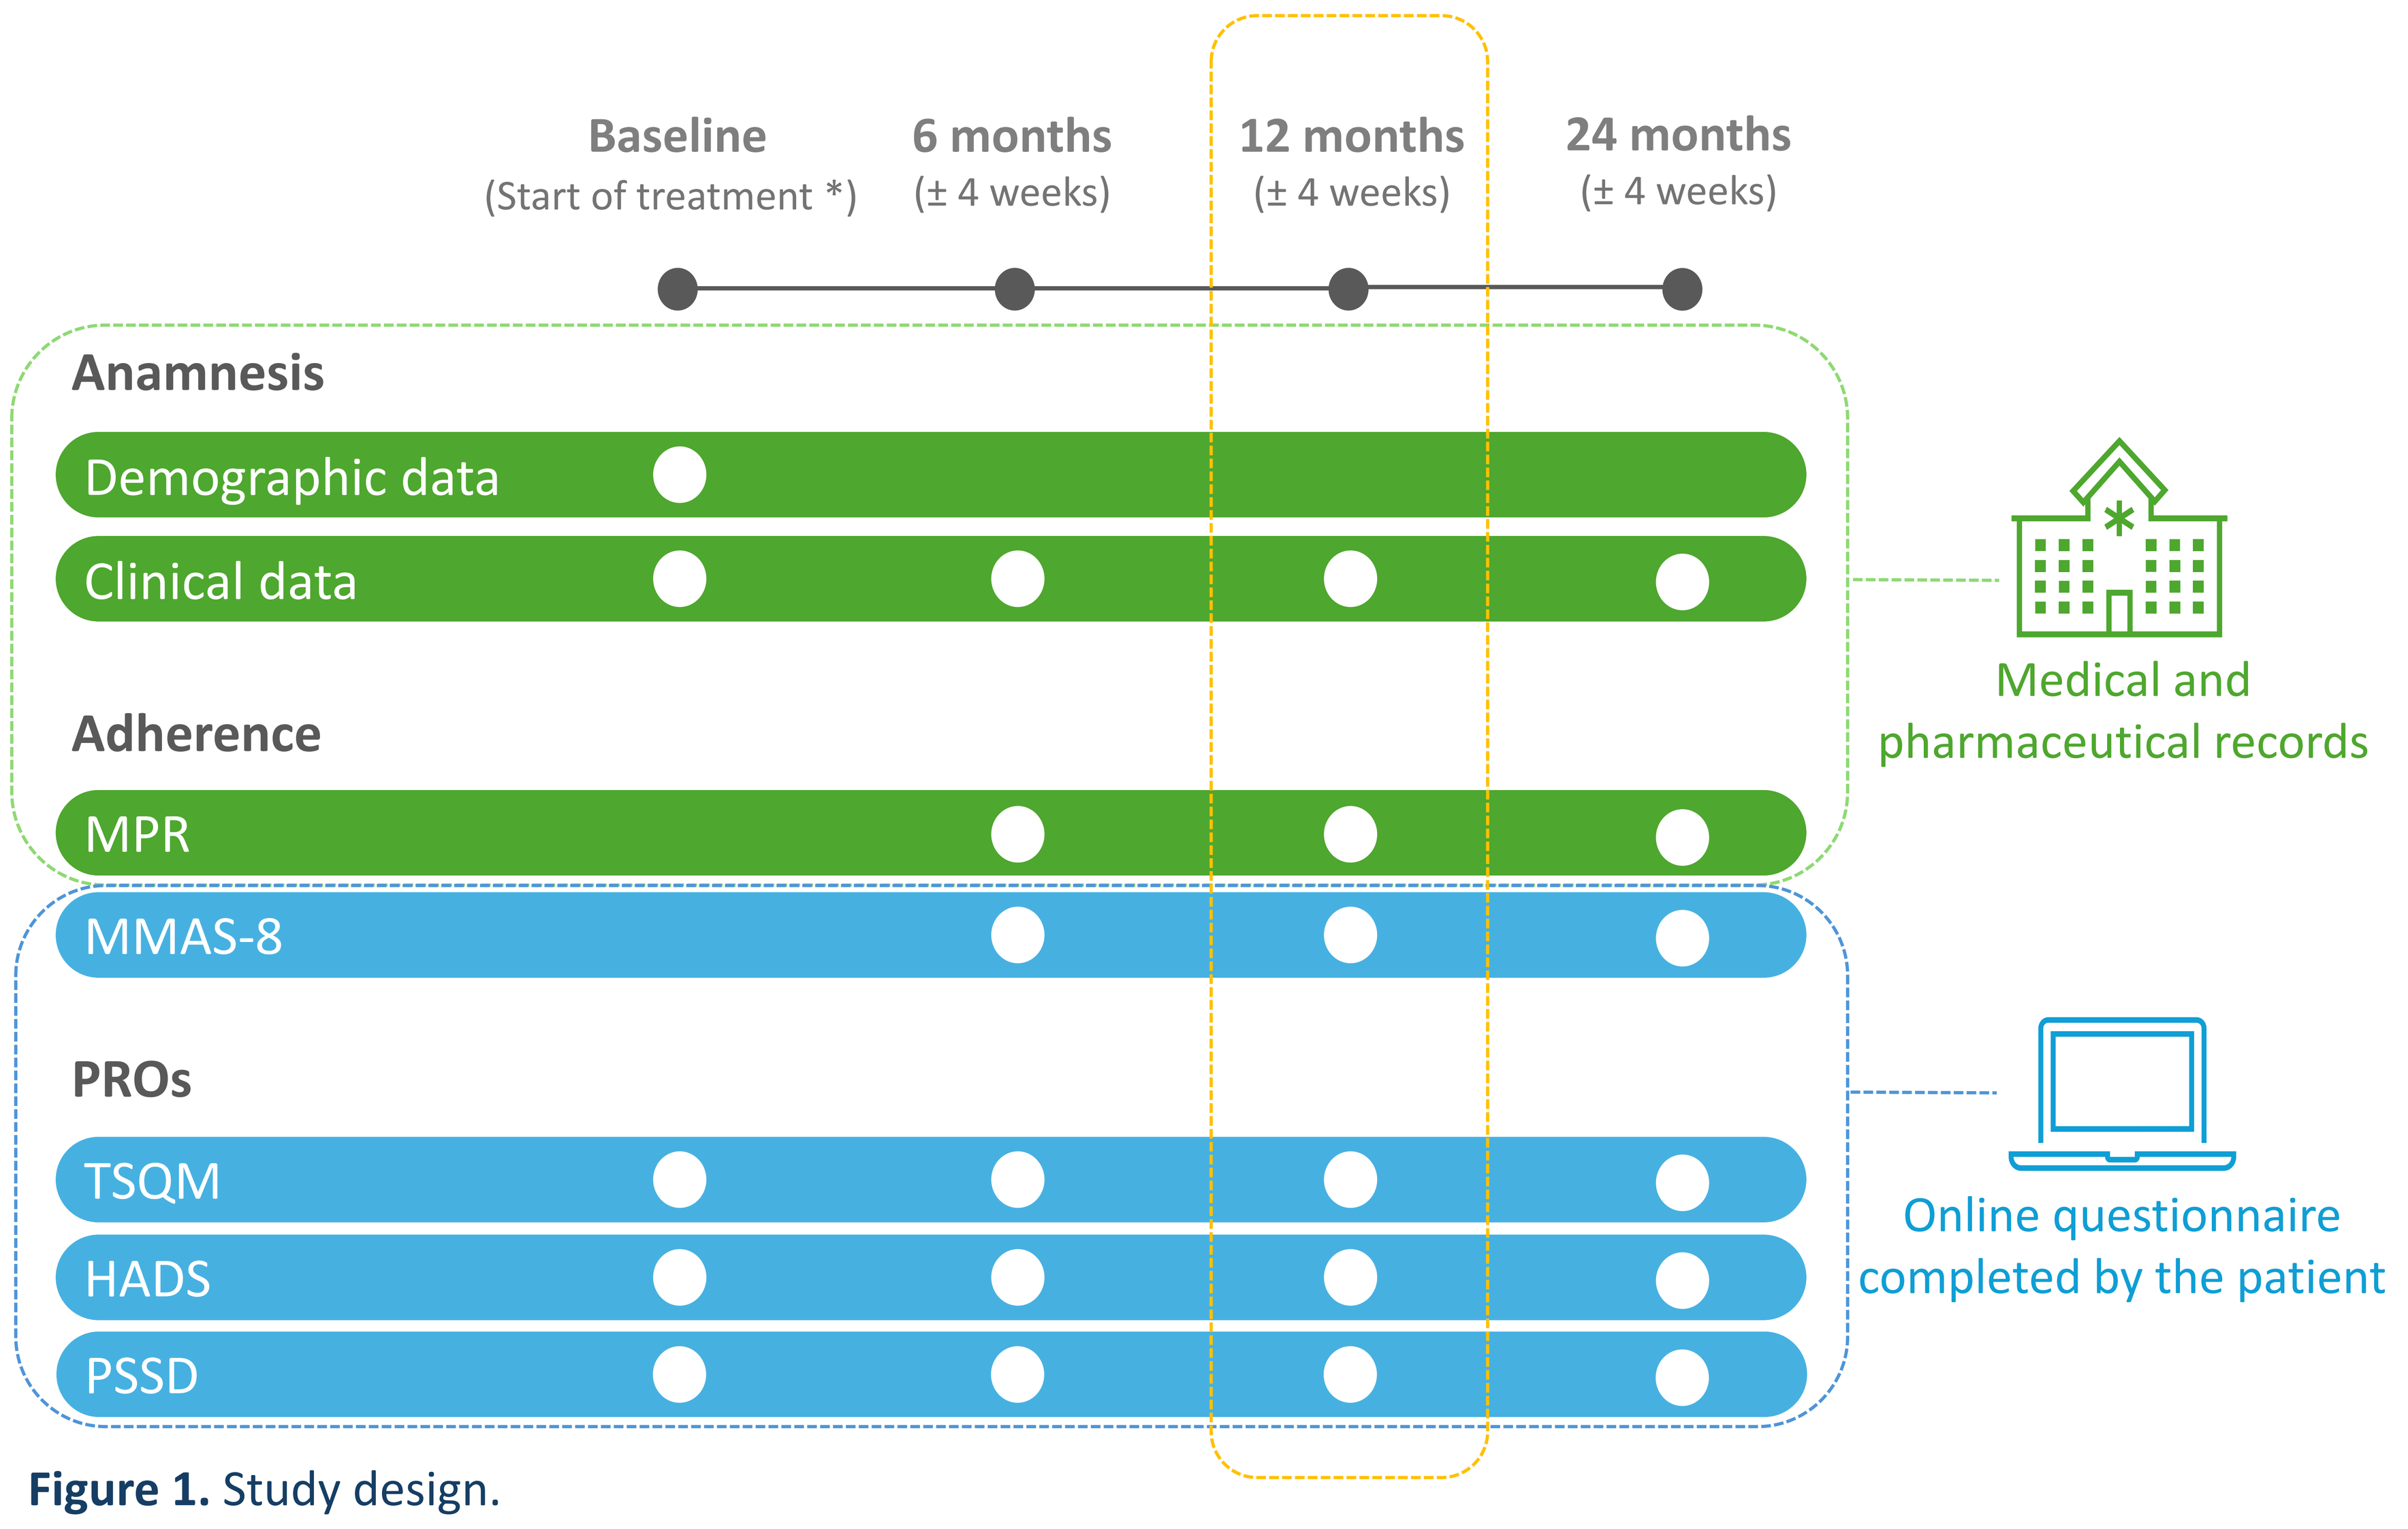
Baseline
(Start of treatment *)
6 months
(± 4 weeks)
12 months
(± 4 weeks)
24 months
(± 4 weeks)
Anamnesis
Demographic data
Clinical data
Adherence
MPR
MMAS-8
PROs
TSQM
HADS
PSSD
Medical and pharmaceutical records
Online questionnaire completed by the patient

Figure 1. Study design.
** Informed consent signature*
***MPR**: Medication Possession Ratio [1]; **MMAS-8**: Morisky Medication Adherence Scale [2]; **PROs**: patient-reported outcomes; **TSQM**: Treatment Satisfaction Questionnaire for Medication[3]; **HADS**: Hospital Anxiety and Depression Scale [4]; **PSSD**: Psoriasis Symptoms and Signs Diary [5];*

Materials and Methods

- Observational, prospective, multicentre study (39 centres) including adult patients with **moderate-to-severe psoriasis** who started **guselkumab** treatment.
- Baseline assessments included **psoriasis area severity index (PASI)**, **body surface area (BSA)** and **dermatology life quality index (DLQI)**.
- We present a **12-month interim analysis** at 09/2025 **(Figure 1).**

Results

- Of 269 patients (mean age 50.8, SD 11.9), 142 (52.8%) completed the 12-month visit.
- At baseline, mean psoriasis duration was 16.5 years (SD 13.5 n=182), 35.3% had psoriatic arthritis, and 81.6% had previously received biologics. Mean basal PASI was 10.1 (SD 7.7, n=193) and mean basal DLQI was 9.8 (SD 6.5, n=107) **(Table 1).**

Table 1. Demographic and clinical data at baseline

Demographic and clinical data		
		N=269
Sex, n (%)	n=269	
Male		145 (53.9)
Female		124 (46.1)
Age (years), mean (SD)	n=269	50.8 (11.5)
BMI (Kg/m^2), mean (SD)	n=256	28.7 (6.1)
Comorbidities, n (%)	n=269	203 (75.5)
Hypertension		73 (27.1)
Dyslipidaemia		88 (32.7)
Psoriatic Arthritis		95 (35.3)
Anxiety/ Depression		62 (23.0)
Time since the first symptoms (years), mean (SD)	n=182	16.5 (13.5)
Bio-experience treatment for psoriasis, n (%)	n=266	217 (81.6)
BSA ≥ 10 %, n (%)	n=211	95 (45.0)
PASI, mean (SD)	n=193	10.1 (7.7)
PASI > 5, n (%)	n=193	137 (71.0)
DLQI, mean (SD)	n=107	9.8 (6.5)

- **At 12 months**, mean MPR was 96.7% (SD 14; range 46–229; n=142). **Adherence** according to MPR≥90%, MMAS-8≥6 and combining criteria is portrayed in **Table 2.**
- **Observed** agreement between MPR≥90% and MMAS-8≥7 was **84.1%** (95/113); There was no systematic difference between methods (McNemar p=0.804); κ=0.123.

Table 2. Patients adherent to treatment according to different criteria

Adherence		n (%)
MPR ≥ 90%	n=142	124 (87.3)
MMAS-8 ≥ 7	n=113	104 (92.0)
Combined		
MPR ≥90% + MMAS-8 ≥7	n=113	95 (84.1)

- **At baseline,** PROs were similar between adherent and non-adherent patients, except for HADS-anxiety (6.6 vs. 10.2; p=0.003) and HADS-depression (5.0 vs. 7.7; p=0.016) **(Figure 2).**
- **At 12 months,** trends favored adherent patients in HADS-anxiety (5.4 vs. 8.7; p=0.002), HADS-depression (4.4 vs. 7.2; p=0.019), PSSD-symptoms (17.2 vs. 31.6; p=0.026) and PSSD-signs (20.3 vs. 33.6; p=0.016) **(Figure 2)**.

Conclusion and Relevance

- **High adherence** (>80%) to **guselkumab** was observed over 12 months treatment.
- Our **preliminary findings** suggest a potential **association** between **adherence** and **improved patient-perceived outcomes**, particularly, psoriasis signs and symptom burden.
- Combining objective and subjective adherence measures may enhance sensitivity in detecting these differences. **These insights** may inform strategies to **optimize long-term psoriasis management.**

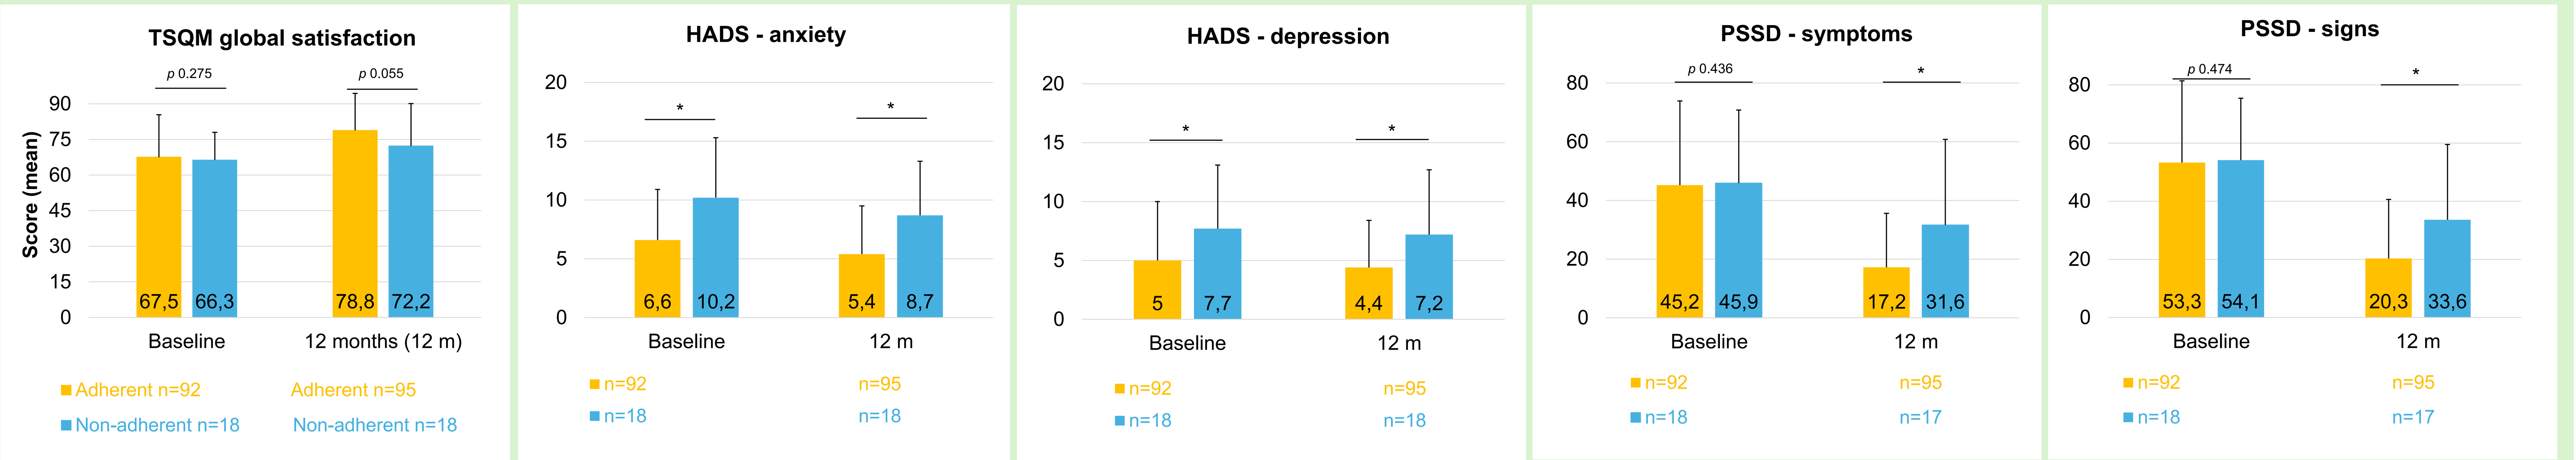
TSQM global satisfaction
Score (mean)
p 0.275
p 0.055
67,5
66,3
78,8
72,2
Baseline
12 months (12 m)
Adherent n=92
Adherent n=95
Non-adherent n=18
Non-adherent n=18
HADS - anxiety
6,6
10,2
5,4
8,7
Baseline
12 m
n=92
n=95
n=18
n=18
HADS - depression
5
7,7
4,4
7,2
Baseline
12 m
n=92
n=95
n=18
n=18
PSSD - symptoms
p 0.436
45,2
45,9
17,2
31,6
Baseline
12 m
n=92
n=95
n=18
n=17
PSSD - signs
p 0.474
53,3
54,1
20,3
33,6
Baseline
12 m
n=92
n=95
n=18
n=17

Figure 2. Mean scores of patient-reported outcomes (TSQM,HADS and PSSD) at baseline and after 12 months of guselkumab treatment in adherent and non-adherent patients, as determined by the combination of MPR and MMAS-8 criteria (MPR ≥ 90% and MMAS-8 ≥ 7). **p* < 0.03 , with Mann–Whitney U test.

References: 1. Sperber, C., *et al.* Patient Prefer. Adherence Volume 11, 1469–1478 (2017). 2. Morisky, D. E., *et al.* J. Clin. Hypertens. 10, 348–354 (2008). 3. Atkinson, M. J. *et al.* Health Qual Life Outcomes 2 (2004). 4. Zigmond, A. S., *et al.* Acta Psychiatr. Scand. 67, 361-370 (1983). 5. Feldman, S. R. *et al.* J. Dermatology Dermatologic Surg. 20, 19–26 (2016).

Acknowledgments: This study was sponsored by Johnson & Johnson. Statistical analyses and study management were performed by Astrum CRO. Medical writing support was provided by Astrum and funded by Johnson & Johnson. We thank the patients and investigators.